Poster # 71

Respiratory Syncytial Virus (RSV) Pre-fusogenic F Nanoparticle Vaccine Compared to Pre-fusion F and Post-fusion F Proteins

G Smith[1], N Patel[1], H Zhou[1], M Massare[1], JH Tian[1], D Scott[1], M Guebre-Xabier[1], H Lu[1], A Portnoff[1], Q Wang[2], Q Xie[2], G Glenn[1]

[1]Novavax, Inc., Gaithersburg, MD; [2]Baylor College of Medicine, Houston, TX

Contact:
Gale Smith, Ph.D.
gsmith@Novavax.com
Phone: 240.268.2000

NOVAVAX
Creating Tomorrow's Vaccines Today

BACKGROUND

Respiratory syncytial virus (RSV) is the leading viral cause of severe lower respiratory tract disease in infants and young children worldwide. RSV fusion (F) envelope glycoprotein is a major target for vaccine development. RSV F is subject to significant rearrangements; precursor (F0) is cleaved by furin forming a fusion inactive F2-p27F1 intermediate (Pre-fusogenic F). A second furin cleavage releases a p27 peptide to form metastable pre-fusion F (Pre-F), which upon rearrangement from a 3-helix to 6-helix bundle post-fusion F configuration (Post-F) drives fusion of viral and cellular membranes. Novavax has developed a near full-length RSV Pre-fusogenic F vaccine composed of purified F2-p27F1 trimers formulated with polysorbate 80 (PS80) as stable 40nm protein – detergent nanoparticles. ResVax (Pre-fusogenic RSV F nanoparticle vaccine) is currently being assessed in the Prepare™ (Phase 3) trial for the protection of infants via maternal immunization in healthy third trimester pregnant women.

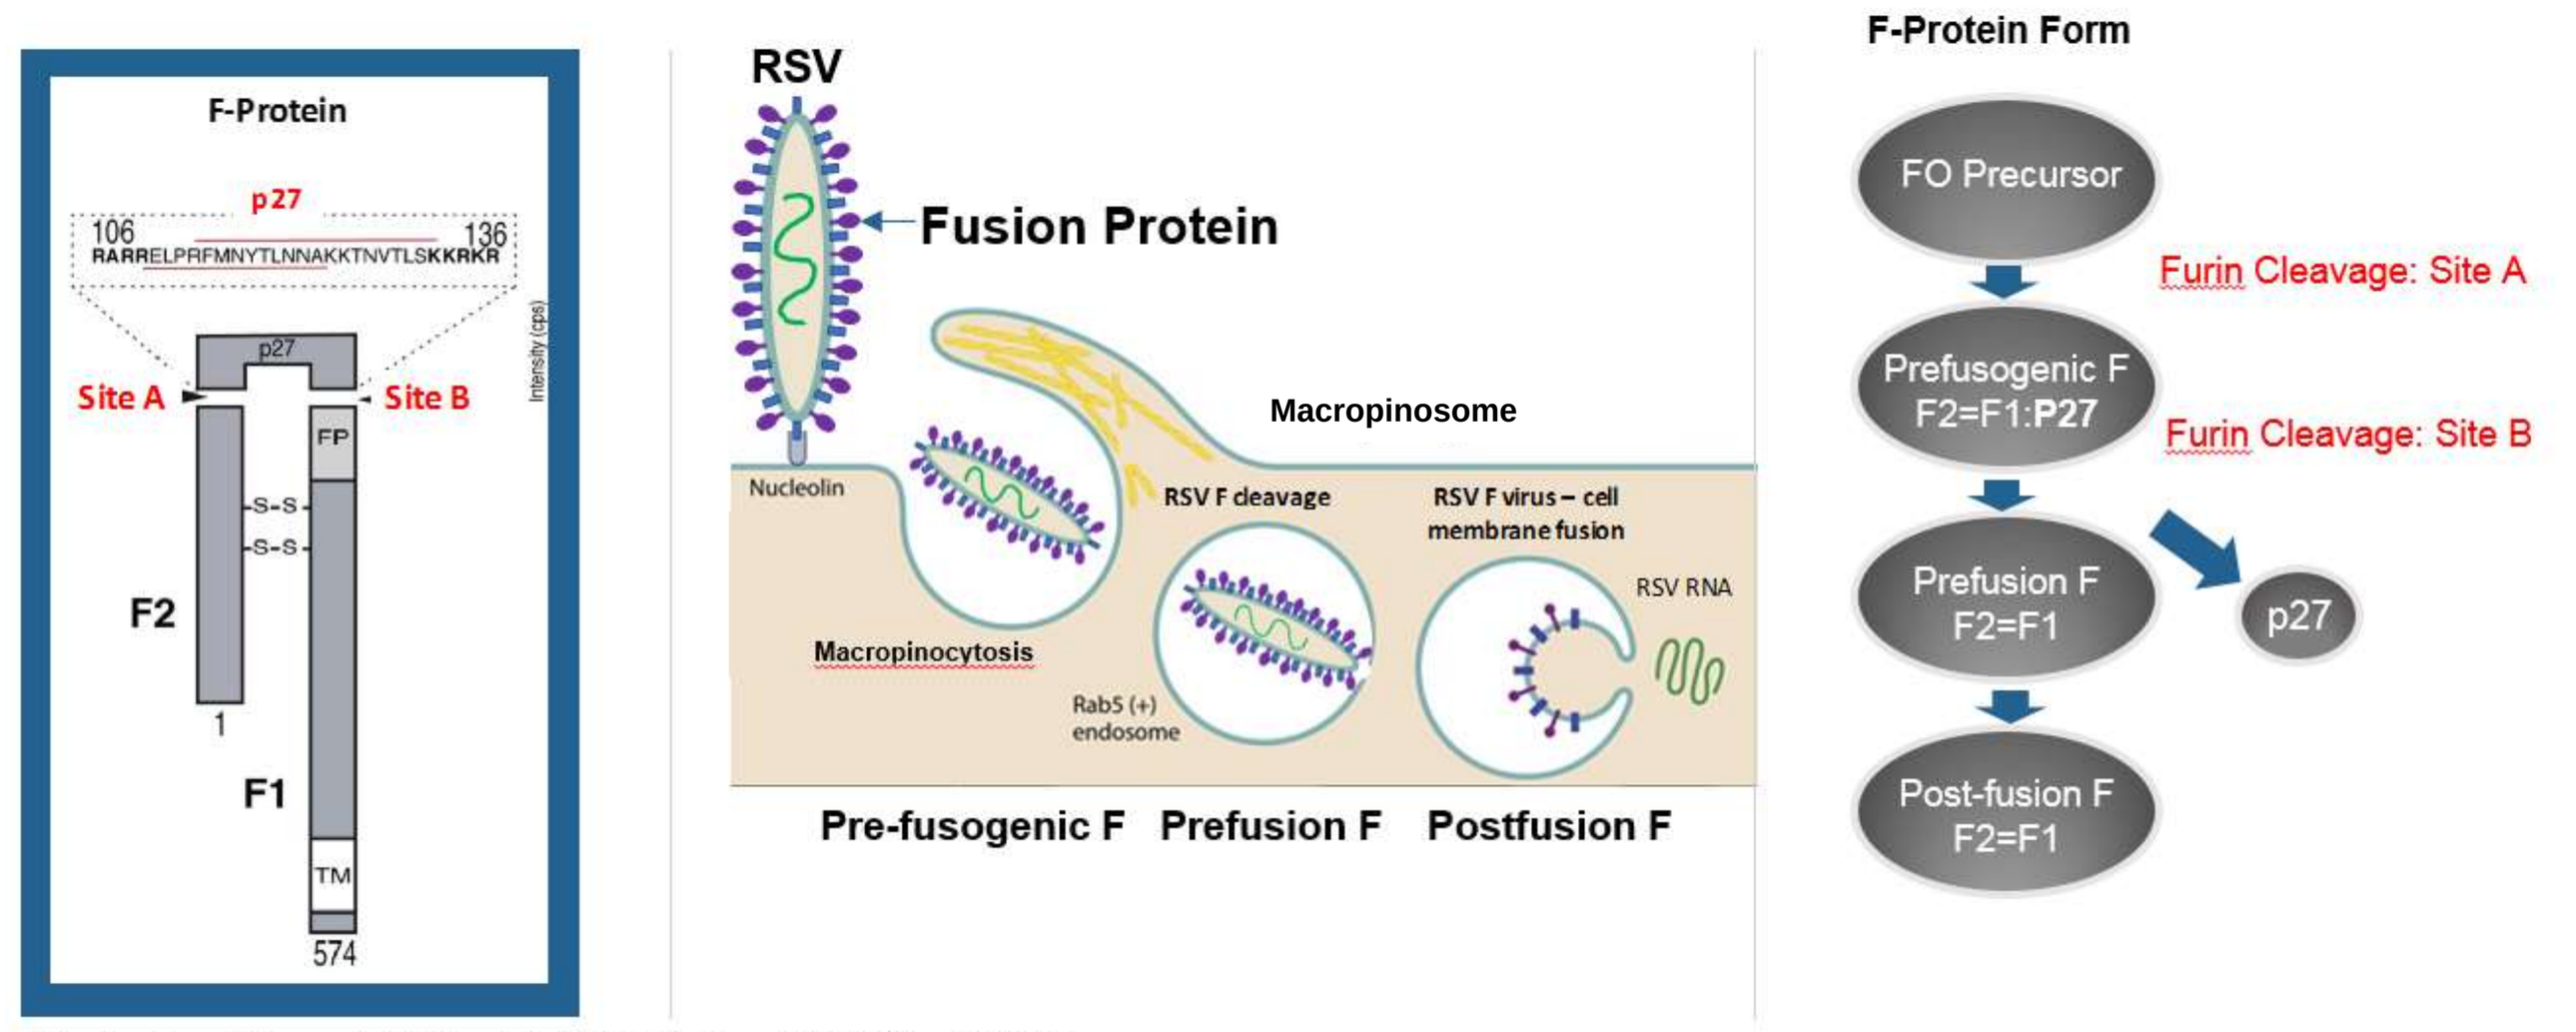
F-Protein
p27
106
136
RARRELPRFMNYTLNNAKKTNVTLSKKRKR
Site A
Site B
FP
F2
F1
TM
574
RSV
Fusion Protein
Macropinosome
Nucleolin
RSV F cleavage
RSV F virus – cell membrane fusion
RSV RNA
Macropinocytosis
Rab5 (+) endosome
Pre-fusogenic F
Prefusion F
Postfusion F
F-Protein Form
FO Precursor
Furin Cleavage: Site A
Prefusogenic F F2=F1:P27
Furin Cleavage: Site B
Prefusion F F2=F1
p27
Post-fusion F F2=F1

Adapted from Krzyzaniak MA, et al. *PLOS Pathog.* 2013;9(4):e1003309:

OBJECTIVES

- Characterization of RSV Pre-fusogenic F nanoparticle vaccine, RSV Pre-F, and Post-F produced in Sf9 cells:
 - Hydrodynamic properties AUC and DLS.
 - Single particle CryoEM at 5 Å resolution.
 - Monoclonal antibody binding to antigenic sites Ø and VIII (pre-fusion specific) and sites II, IV, II/IV, and p27.
- FACS analyses of recombinant, membrane associated RSV F:
 - Insect Sf9 cells: RSV Pre-fusogenic F and Pre-F.
 - Human 293T: RSV Pre-fusogenic F and wild type F.
- RSV Pre-fusogenic, Pre-F, and Post-F immunogenicity in mice.

METHODS

- RSV Pre-fusogenic F (GMP drug substance), Pre-F (single-chain triple mutant; SCTM), and Post-F were produced in Sf9 cells.
- Hydrodynamic properties purified RSV F antigens were measured using analytical ultracentrifugation (AUC) and dynamic light scattering (DLS).
- Single particle CryoEM of a 72-aa C-terminal truncated Pre-fusogenic F was compared to Pre-F and Post-F at 5 Å resolution.
- Antigen binding of mAbs specific for pre-fusion sites Ø and VIII and sites II, IV, II/IV and p27 using Octet 384 Bio-Layer Interferometry and dot blot.
- FACS cell surface expressed RSV Pre-fusogenic F and Pre-F on Sf9 cells and Pre-fusogenic F and wild type RSV/A2 F on human 293T cells.
- Immunogenicity 1µg and 10µg antigens w/wo AlPO4 adjuvant in mice.

RESULTS

Fig 1. **RSV Pre-fusogenic, Pre-F, Post-F Proteins**

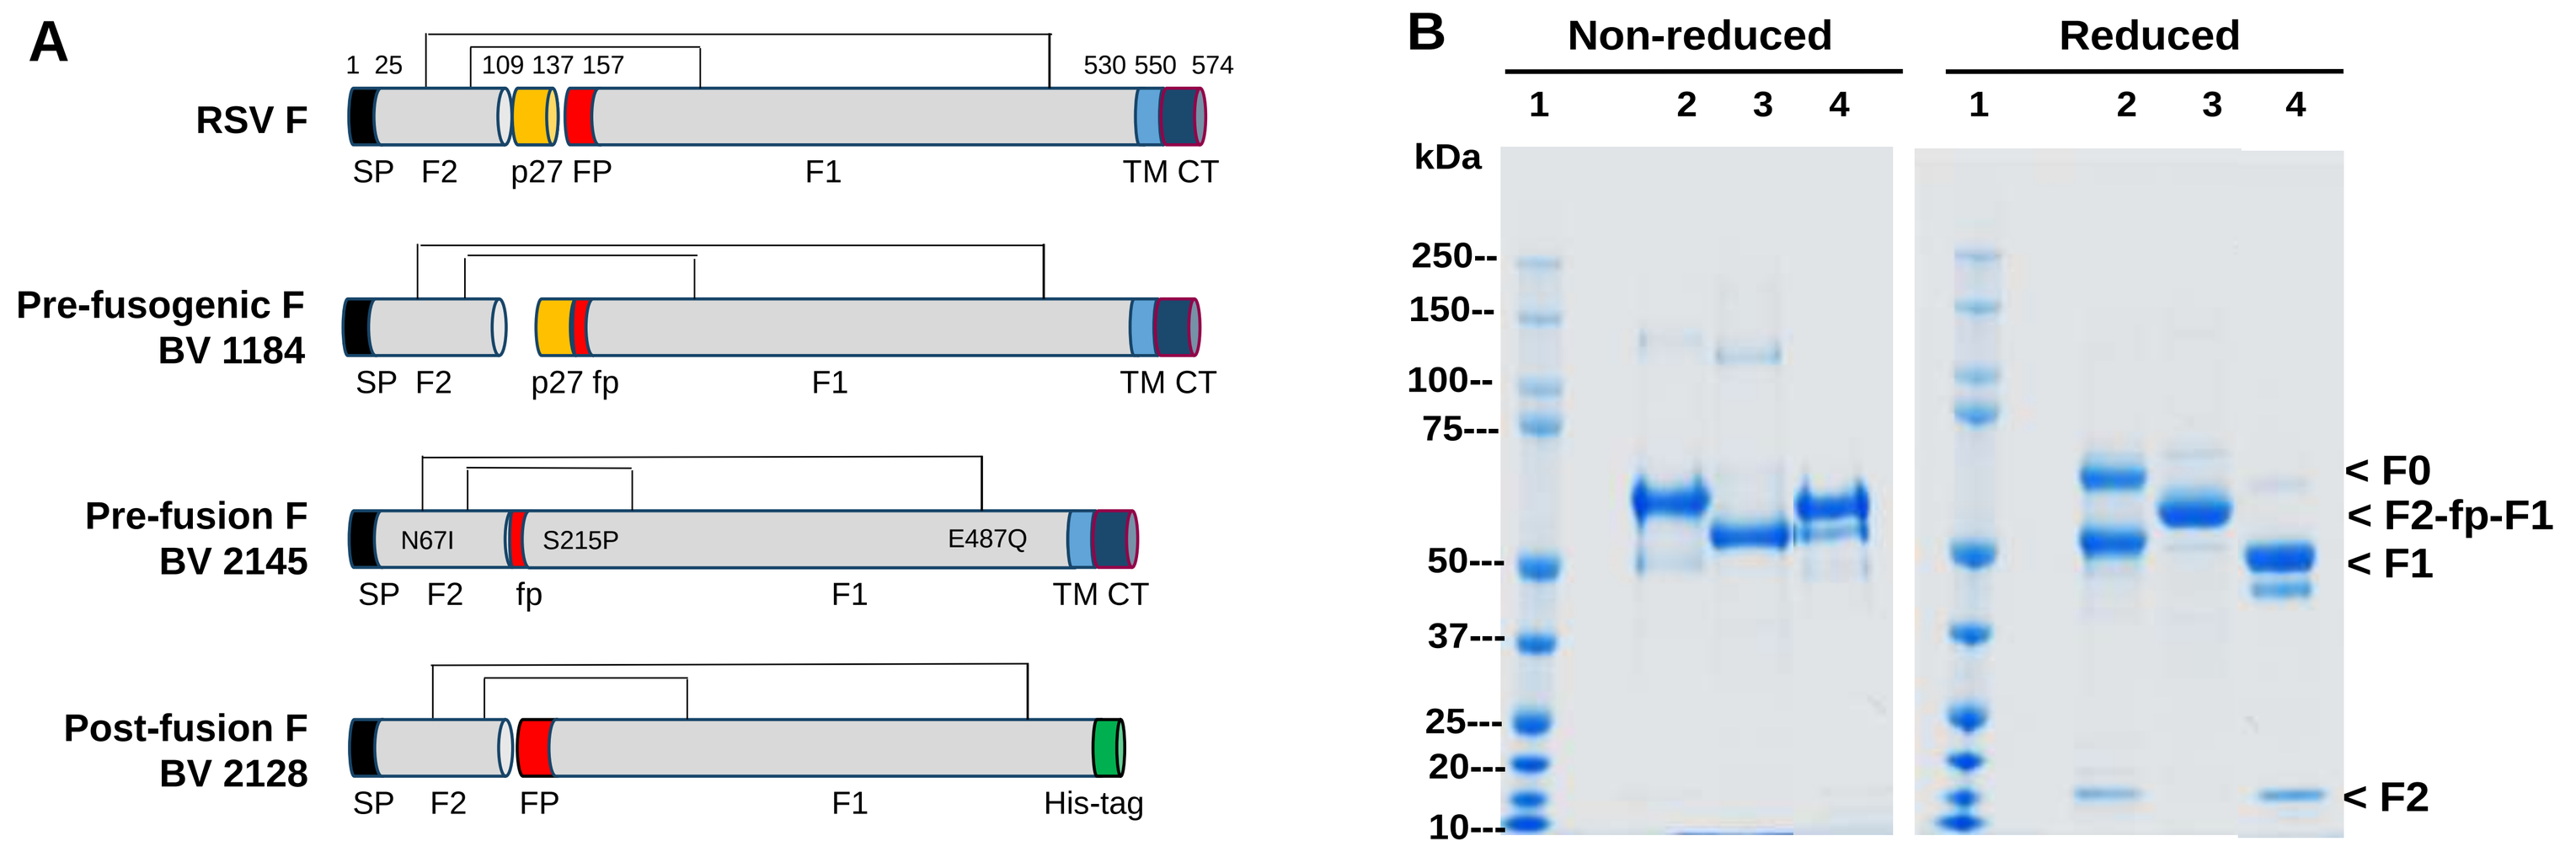
A
RSV F
1 25
109 137 157
530 550 574
SP F2 p27 FP F1 TM CT
Pre-fusogenic F BV 1184
SP F2 p27 fp F1 TM CT
Pre-fusion F BV 2145
N67I S215P E487Q
SP F2 fp F1 TM CT
Post-fusion F BV 2128
SP F2 FP F1 His-tag
B
Non-reduced
Reduced
1 2 3 4
kDa
250--
150--
100--
75---
50---
37---
25---
20---
10---
< F0
< F2-fp-F1
< F1
< F2

Fig 2. **Hydrodynamic parameters RSV F constructs: AUC and DLS**

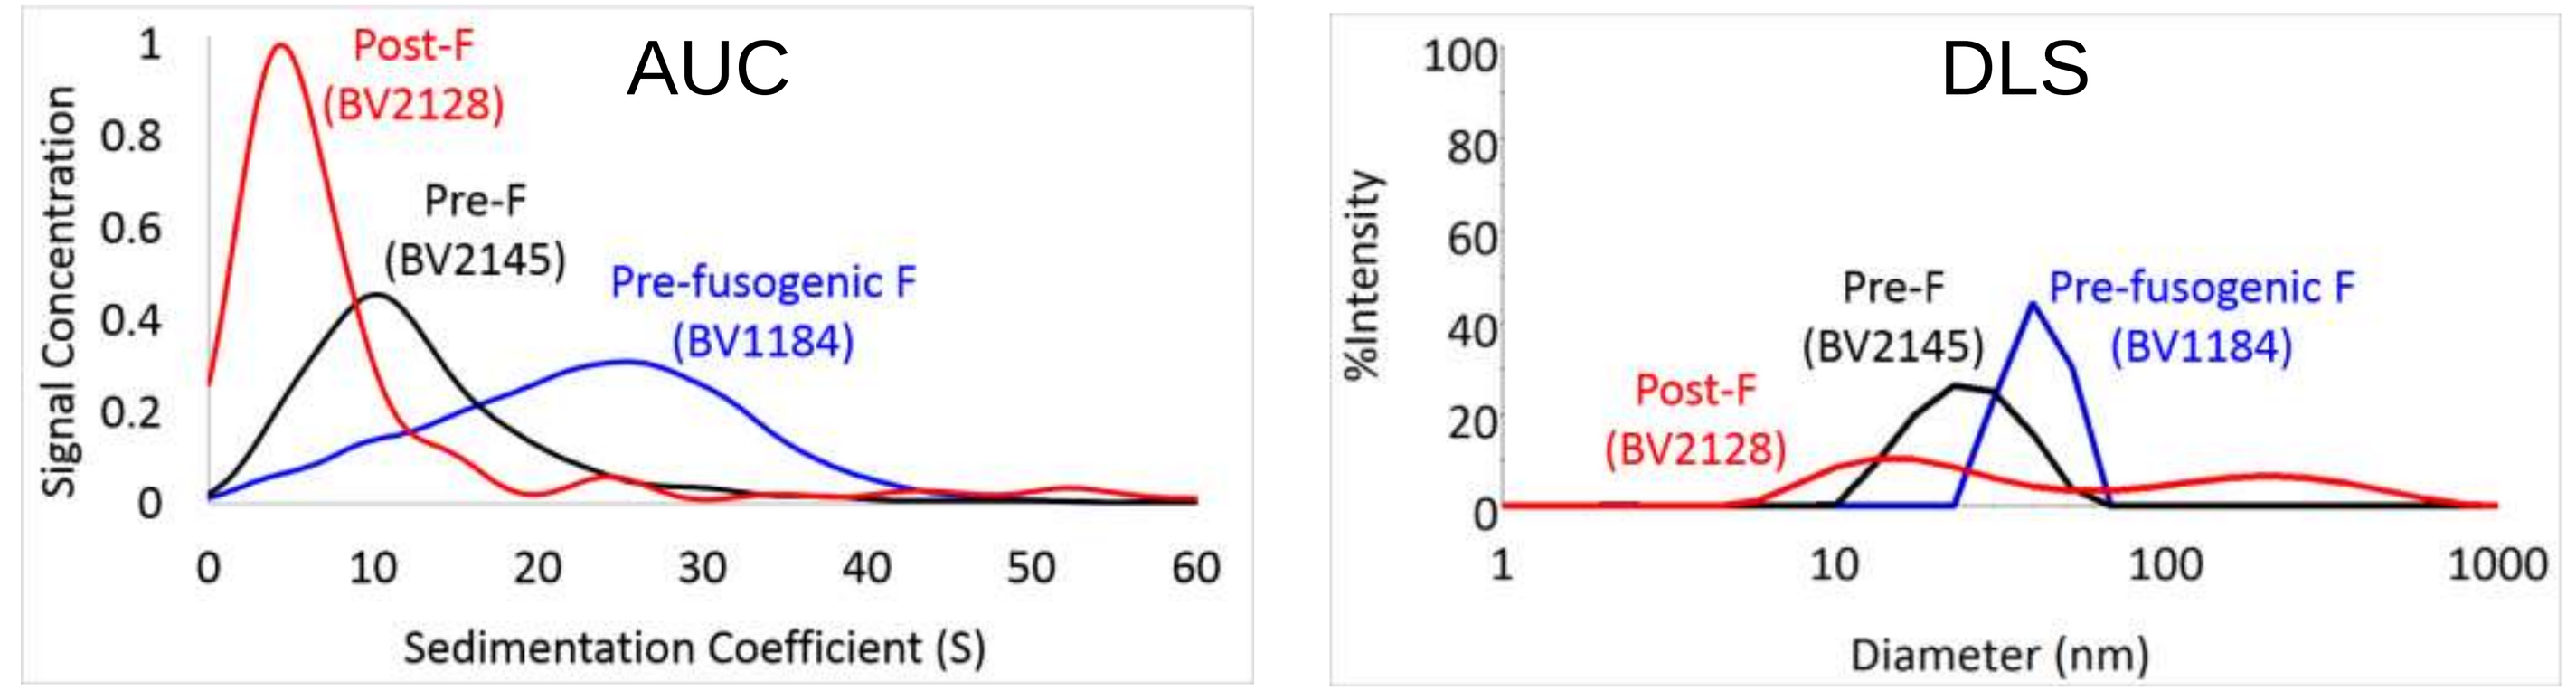
AUC
Post-F (BV2128)
Pre-F (BV2145)
Pre-fusogenic F (BV1184)
Signal Concentration
Sedimentation Coefficient (S)
DLS
%Intensity
Diameter (nm)

Hydrodynamic parameters of RSV fusion glycoprotein constructs

RSV F proteins	SV-AUC: $s^0_{20°C,w}$, Svedberg	SV-AUC: Mw, app kDa²	SV-AUC: f/f_0 $_{20°C,w}$	DLS: Z-average (nm)	DLS: PDI
Pre-fusogenic F	23.3 S	1,146	1.11	40	0.17
Pre-fusion F	13.7 S	710	1.12	23	0.15
Post-fusion F	5.0 S	123	1.24	28	Multi-modal

Fig 3. **Single Particle CryoEM RSV Pre-fusogenic F 5 Å Resolution**

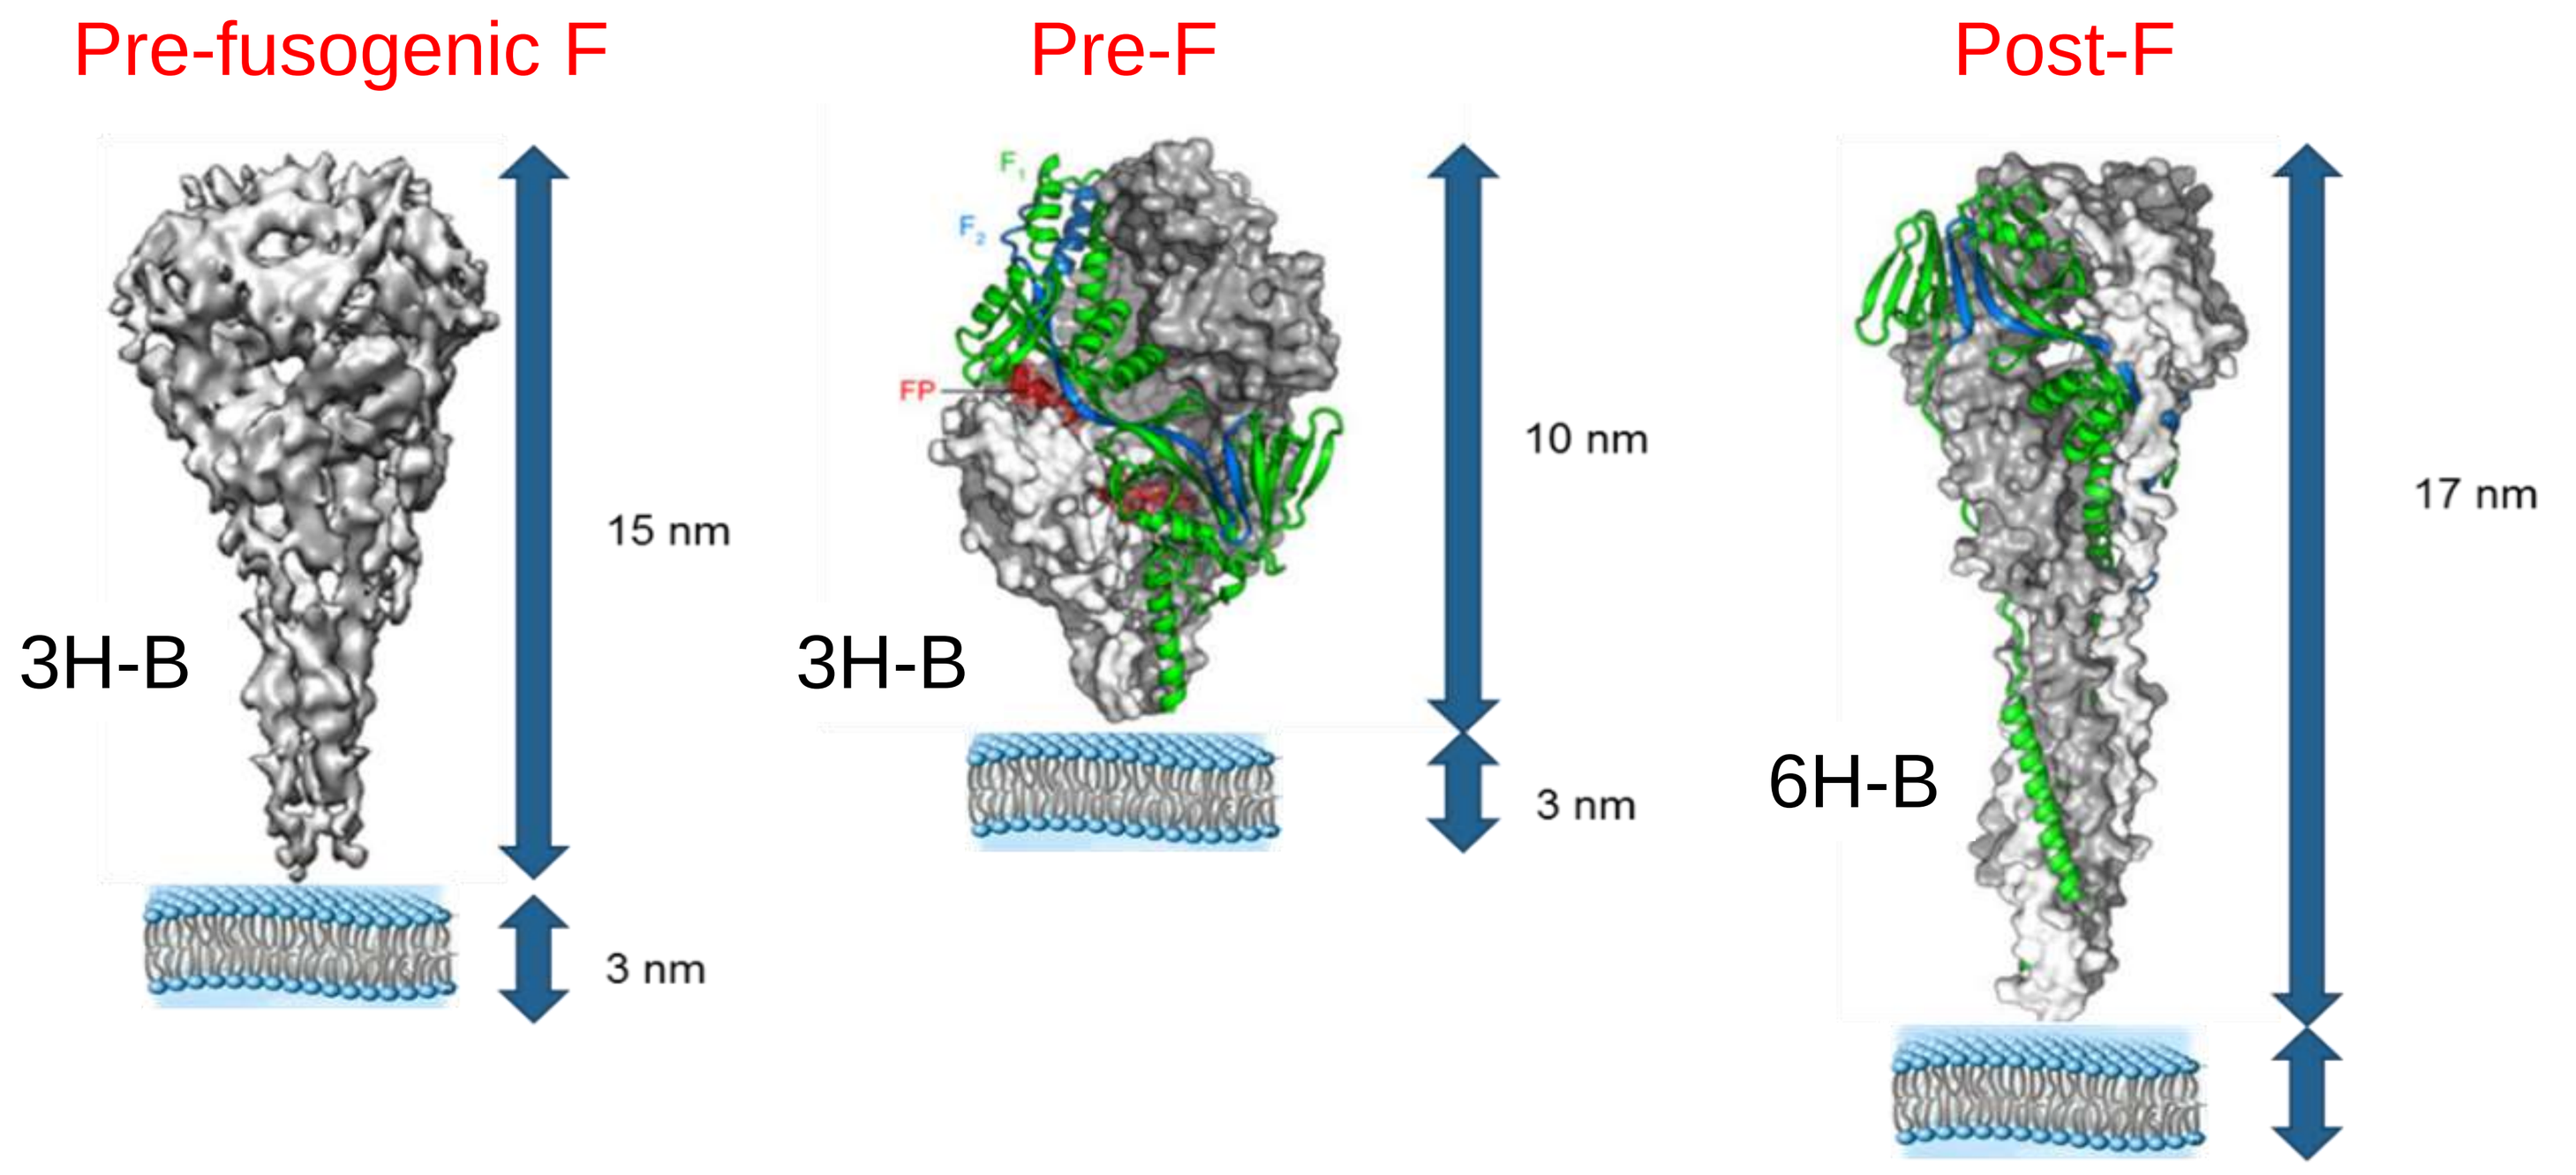
Pre-fusogenic F
Pre-F
Post-F
3H-B
15 nm
3 nm
3H-B
10 nm
3 nm
6H-B
17 nm
FP

Fig 4. **Bio-layer interferometry and dot blot of RSV F mAb binding**

Octet Bio-layer Interferometry

Antigenic Site	Monoclonal Antibody	% Binding Monoclonal Antibody: Pre-fusogenic F	Pre-F	Post-F
Site Ø	D25	40%	126%	0%
	hRSV106	15%	104%	0%
Site VIII	hRSV90	39%	149%	0%
Site II	Palivizumab	113%	126%	108%
Site IV	RSHZ19	104%	112%	107%
	R1.42	95%	89%	77%
Site II/IV	R4.C6	94%	85%	78%
P27	RSV.7.10	91%	0%	0%

No binding / Intermediate binding / High binding

Dot blot

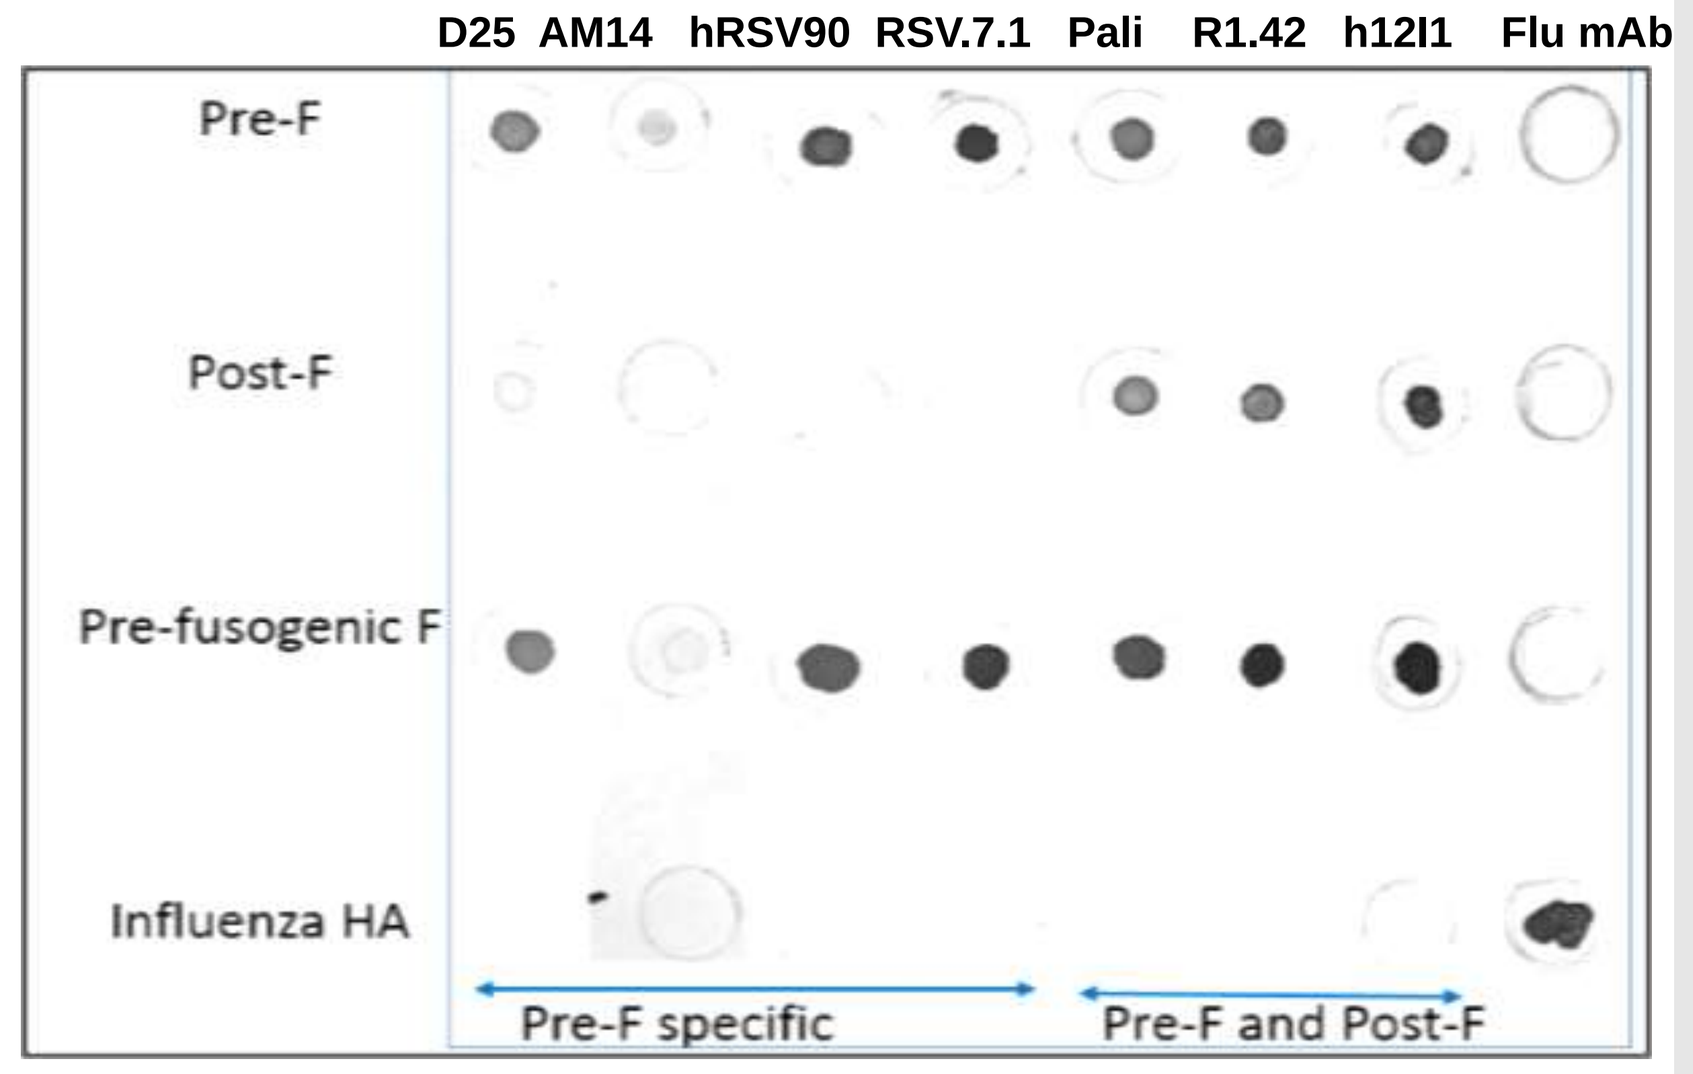
D25 AM14 hRSV90 RSV.7.1 Pali R1.42 h12I1 Flu mAb
Pre-F
Post-F
Pre-fusogenic F
Influenza HA
Pre-F specific
Pre-F and Post-F

RESULTS

Fig 5. **A. FACS Pre-fusogenic F and Pre-F Sf9 cells**
B. FACS RSV Pre-fusogenic F or wild-type F hu293T cells

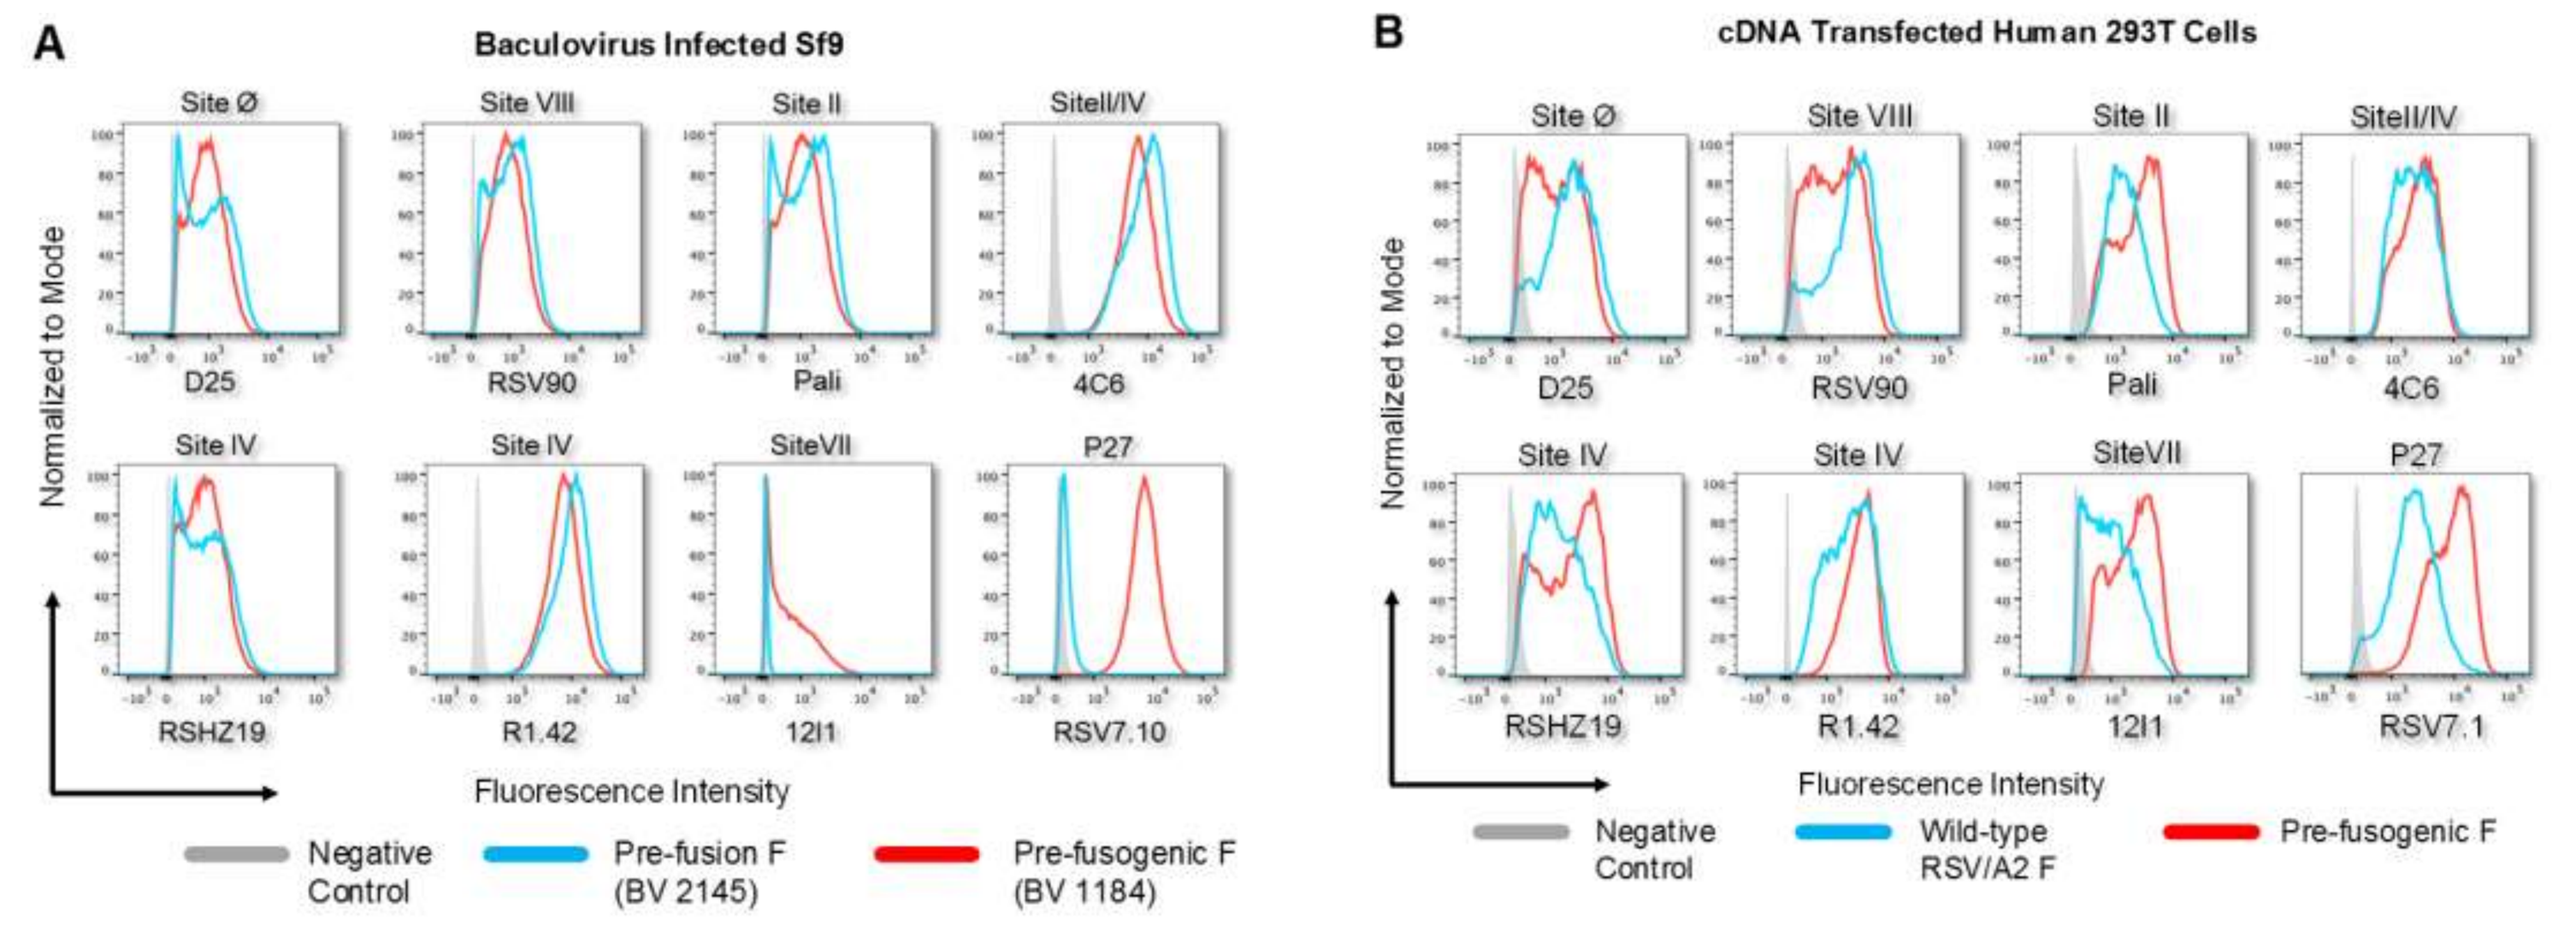
A
Baculovirus Infected Sf9
B
cDNA Transfected Human 293T Cells
Site Ø
Site VIII
Site II
SiteII/IV
D25
RSV90
Pali
4C6
Site IV
SiteVII
P27
RSHZ19
R1.42
12I1
RSV7.10
RSV7.1
Normalized to Mode
Fluorescence Intensity
Negative Control
Pre-fusion F (BV 2145)
Pre-fusogenic F (BV 1184)
Wild-type RSV/A2 F
Pre-fusogenic F

Fig 6. **RSV F ELISA and Microneutralization responses in mice**

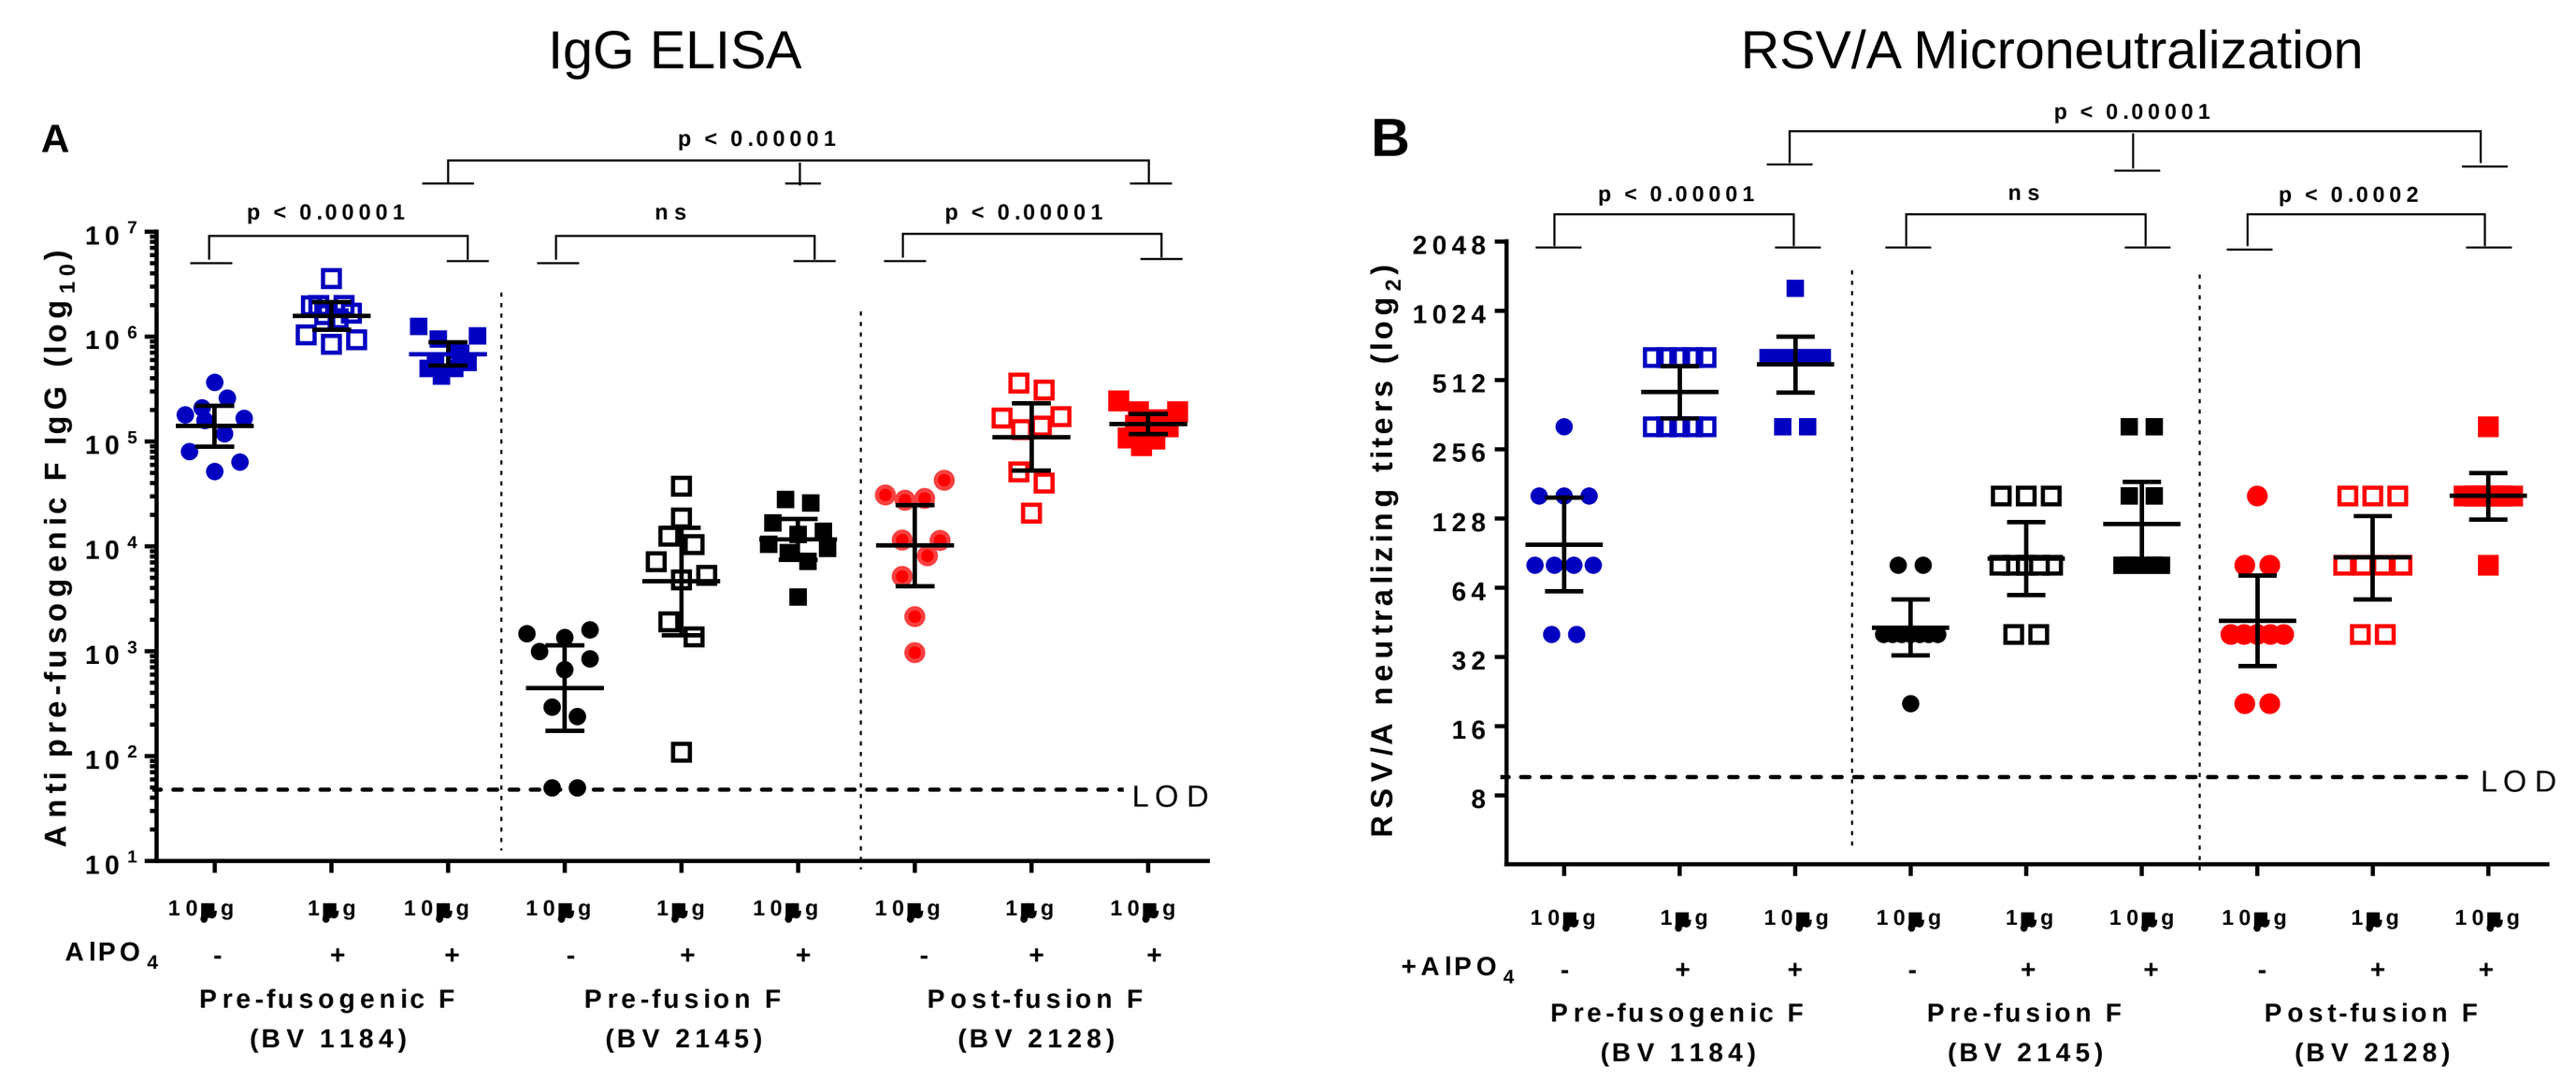
IgG ELISA
RSV/A Microneutralization
A
B
p < 0.00001
ns
p < 0.0002
Anti pre-fusogenic F IgG (log10)
RSV/A neutralizing titers (log2)
LOD
AlPO4
Pre-fusogenic F (BV 1184)
Pre-fusion F (BV 2145)
Post-fusion F (BV 2128)

CONCLUSIONS

- Pre-fusogenic F forms a distinct protein – PS80 detergent nanoparticle.
- Pre-fusogenic F with a 72aa truncation of the C-terminus form trimers with a 3helix-bundle stem and a length intermediate between Pre-F and Post-F.
- Pre-fusogenic F expressed on Sf9 and human 293T cells and when purified bind pre-fusion-specific, pre/post-fusion, and p27 monoclonal antibodies.
- Pre-fusogenic F nanoparticle vaccine without and with $AlPO_4$ adjuvant was significantly more immunogenic in mice than Pre-F and Post-F.

REFERENCES

1. Smith G, et al. Respiratory syncytial virus fusion glycoprotein expressed in insect cells form protein nanoparticles that induce protective immunity in cotton rats. PLOS ONE 2012;7:e50852.
2. Krarup A, et al. A highly stable prefusion RSV F vaccine derived from structural analysis of the fusion mechanism. 2015. Nature Communications. DOI:10.1038/ncomms9143.
3. McLellan JS. Neutralizing epitopes on the respiratory syncytial virus fusion glycoprotein. Current Opinion in Virology 2015, 11:70-75.
4. Swanson KA, et al. Structural basis for immunization with postfusion respiratory syncytial virus fusion F glycoprotein (RSV F) to elicit high neutralizing antibody titers. 2011. PNAS. DOI.10.1073/pnas.1106536108.

Conflict of Interest Disclosure: GS, NP, HZ, MM, JHT, DS, MGX, HL, AP, and GG are employees of Novavax, Inc. QW and QX are employees of Baylor College of Medicine. The authors have no other relevant affiliations or financial involvement with any organization or entity with a financial interest in or financial conflict with the subject matter of materials discussed in the poster apart from those disclosed.